Ongoing Natural History Study in Phosphomannomutase 2 Congenital Disorder of Glycosylation (PMM2-CDG): Clinical and Basic Investigations

Eva Morava[1], Marc Patterson[1], Anna Čechová[2], Tomáš Honzík[2], Andrew C. Edmondson[3], Can Ficicioglu[3], Mercedes Serrano[4], Rita Barone[5], Peter Witters[6], Daisy Ryman[6], Christina Lam[7], Mirian C.H. Janssen[8], Esmeralda Martins[9], Dulce Quelhas[9], Jolanta Sykut-Cegielska[10], Claire-Marine Bérat[11], Pascale De Lonlay[11]

1. Department of Clinical Genomics-Department of Laboratory Medicine and Pathology, Mayo Clinic, MN, USA 2. Department of Pediatrics and Inherited Metabolic Disorders, First Faculty of Medicine, Charles University and General University Hospital in Prague, Prague, Czech Republic 3. Division of Human Genetics, Department of Pediatrics, Children's Hospital of Philadelphia, USA 4. Pediatric Neurology Department, Hospital Sant Joan de Déu, Institut de Recerca Sant Joan de Déu, Barcelona, Spain 5. Child Neuropsychiatry Unit, Department of Clinical and Experimental Medicine, University of Catania, Catania, Italy 6. Metabolic Center, Department of Pediatrics, University Hospitals Leuven, Herestraat 49, 3000 Leuven, Belgium 7. Division of Genetic Medicine, Department of Pediatrics, University of Washington School of Medicine, Seattle, WA, USA 8. Radboud University Medical Centre, Department of Internal Medicine, Nijmegen, the Netherlands 9. Centro Hospitalar Universitário do Porto, Porto, Portugal 10. Department of Inborn Errors of Metabolism and Paediatrics, the Institute of Mother and Child, Warsaw, Poland 11. Necker Hospital, APHP, Reference Center for Inborn Errors of Metabolism, University of Paris, Paris, France

Abstract

GLY-000 Natural History study (NCT03173300) is an ongoing international, multi-center study being conducted at 11 sites, 8 in Europe and 3 in the USA. The goal of the study is to collect cross-sectional and longitudinal data on clinical and biochemical parameters to inform the design of future interventional studies. Several key findings have been observed.

- ***Failure to thrive:*** most acute in infants, persists throughout life
- ***Coagulation parameters:*** dysregulated across a majority of patients, persisting into adulthood; no trend over study duration, with visit-to-visit variability
- ***Carbohydrate deficient transferrin:*** shows no trend over study duration, but visit-to-visit variability
- ***Liver enzymes:*** elevated in children, particularly those < 6 years of age
- ***Adrenal insufficiency:*** ~ 25% of patients; screening now incorporated into standard-of-care

Background

PMM2-CDG is the most common CDG, representing ~65% of cases. The PMM2 enzyme converts mannose-1-phosphate (M1P) to mannose-6-phosphate (M6P). A deficiency in PMM2 results in hypo-glycosylated proteins and drives the pathophysiology of PMM2-CDG. Clinical presentation is variable, ranging from infants who die in the first weeks of life, to mildly affected adults. Apart from neurologic manifestations, patients have variable, sometimes severe, immunologic, hematologic, cardiac, pulmonary, gastrointestinal, renal, endocrine, musculoskeletal, developmental and/or ophthalmologic involvement.

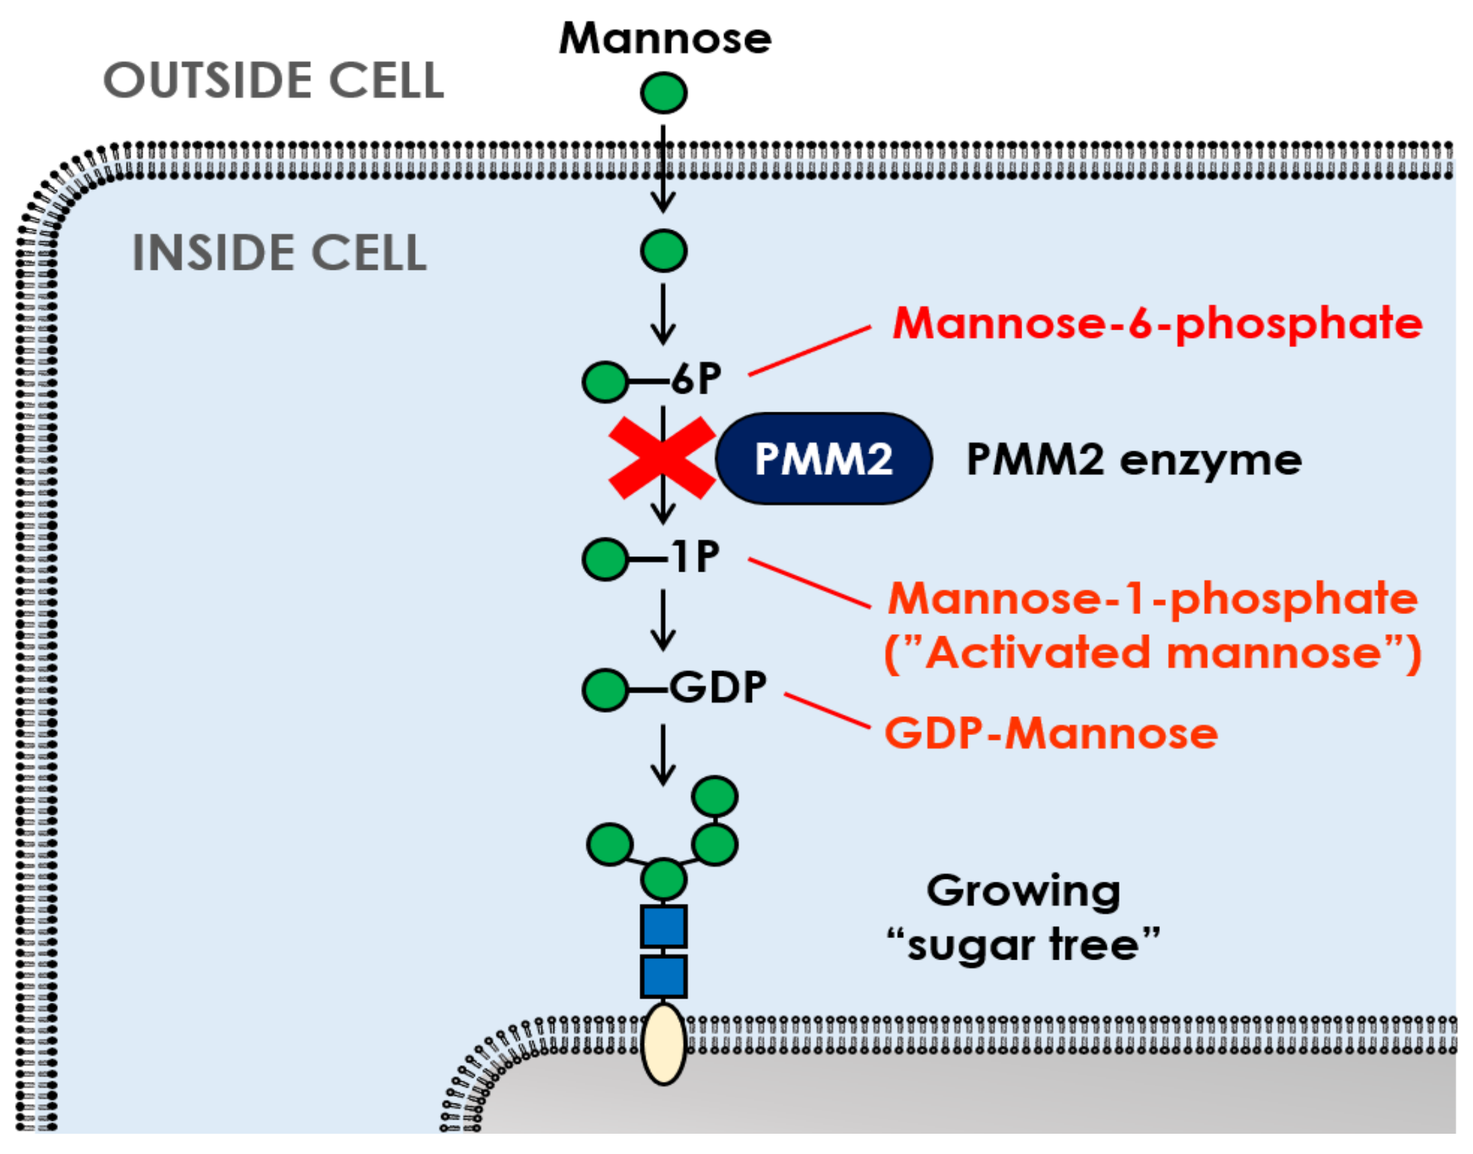

Mannose
OUTSIDE CELL
INSIDE CELL
6P
Mannose-6-phosphate
PMM2
PMM2 enzyme
1P
Mannose-1-phosphate
("Activated mannose")
GDP
GDP-Mannose
Growing
"sugar tree"

Results

Demographics

A total of 139 participants have been enrolled in the study, with an average age of 10.4 yrs (0.5 to 68.4) at baseline. All age groups are well represented, with the largest cohort (32%) in the 0 to 6 yrs age range, followed by adults > 18 yrs (31%). Males (53%) and females (47%) are approximately equally represented.

Demographic	Statistic	Value
Age (years)	n	139
	Median (Max, Min)	10.40 (0.5, 68.4)
Age Group (years)	0 - <6 years	44 (31.7%)
	6 - <12 years	33 (23.7%)
	12 - <18 years	19 (13.7%)
	>= 18 years	43 (30.9%)
Sex	Female	65 (46.8%)
	Male	74 (53.2%)

Enrollment

Enrollment in the study took place over approximately 3 years, beginning in March 2018. Currently 16 (11.5%) participants have completed 4 years on study and all remaining patients have completed at least 2 years on study. There have been 13 discontinuations, including 3 deaths; 4 yrs/F (H1N1 viral pneumonia, kidney failure), 2 yrs/F (systemic infection), and 43 yrs/M (respiratory failure).

Degree of Completion	Overall (N=139)
0 Years	3 (2.2%)
1 Year	7 (5.0%)
2 Years	71 (51.1%)
3 Years	42 (30.2%)
4 Years	16 (11.5%)
Discontinuations	**Overall (N=13)**
Death	3 (2.1%)
Withdrew Consent	4 (2.8%)
Administrative / Other	6 (4.2%)

Genotypes

Of the139 individuals enrolled,137 participants had complete genotype information. A total of 60 unique variants were found. The majority (n=80, 58.4%) had the **R141H** variant, in the substrate binding domain, with the next most frequent, **P113L** (n=29, 21.2%) and **F119L** (n=17, 12.4%), in the dimerization domain of the enzyme.

Variant Frequencies (Top 10; in >5% pts)

Variant	All (N=137)
R141H	80 (58.4%)
P113L	29 (21.2%)
F119L	17 (12.4%)
V231M	11 (8.0%)
C241S	11 (8.0%)
F157S	9 (6.6%)
R162W	9 (6.6%)
T237M	9 (6.6%)
T237R	8 (5.8%)
V129M	7 (5.1%)

Variant Pair (Heterozygous) Frequencies (> 3 occurrences)

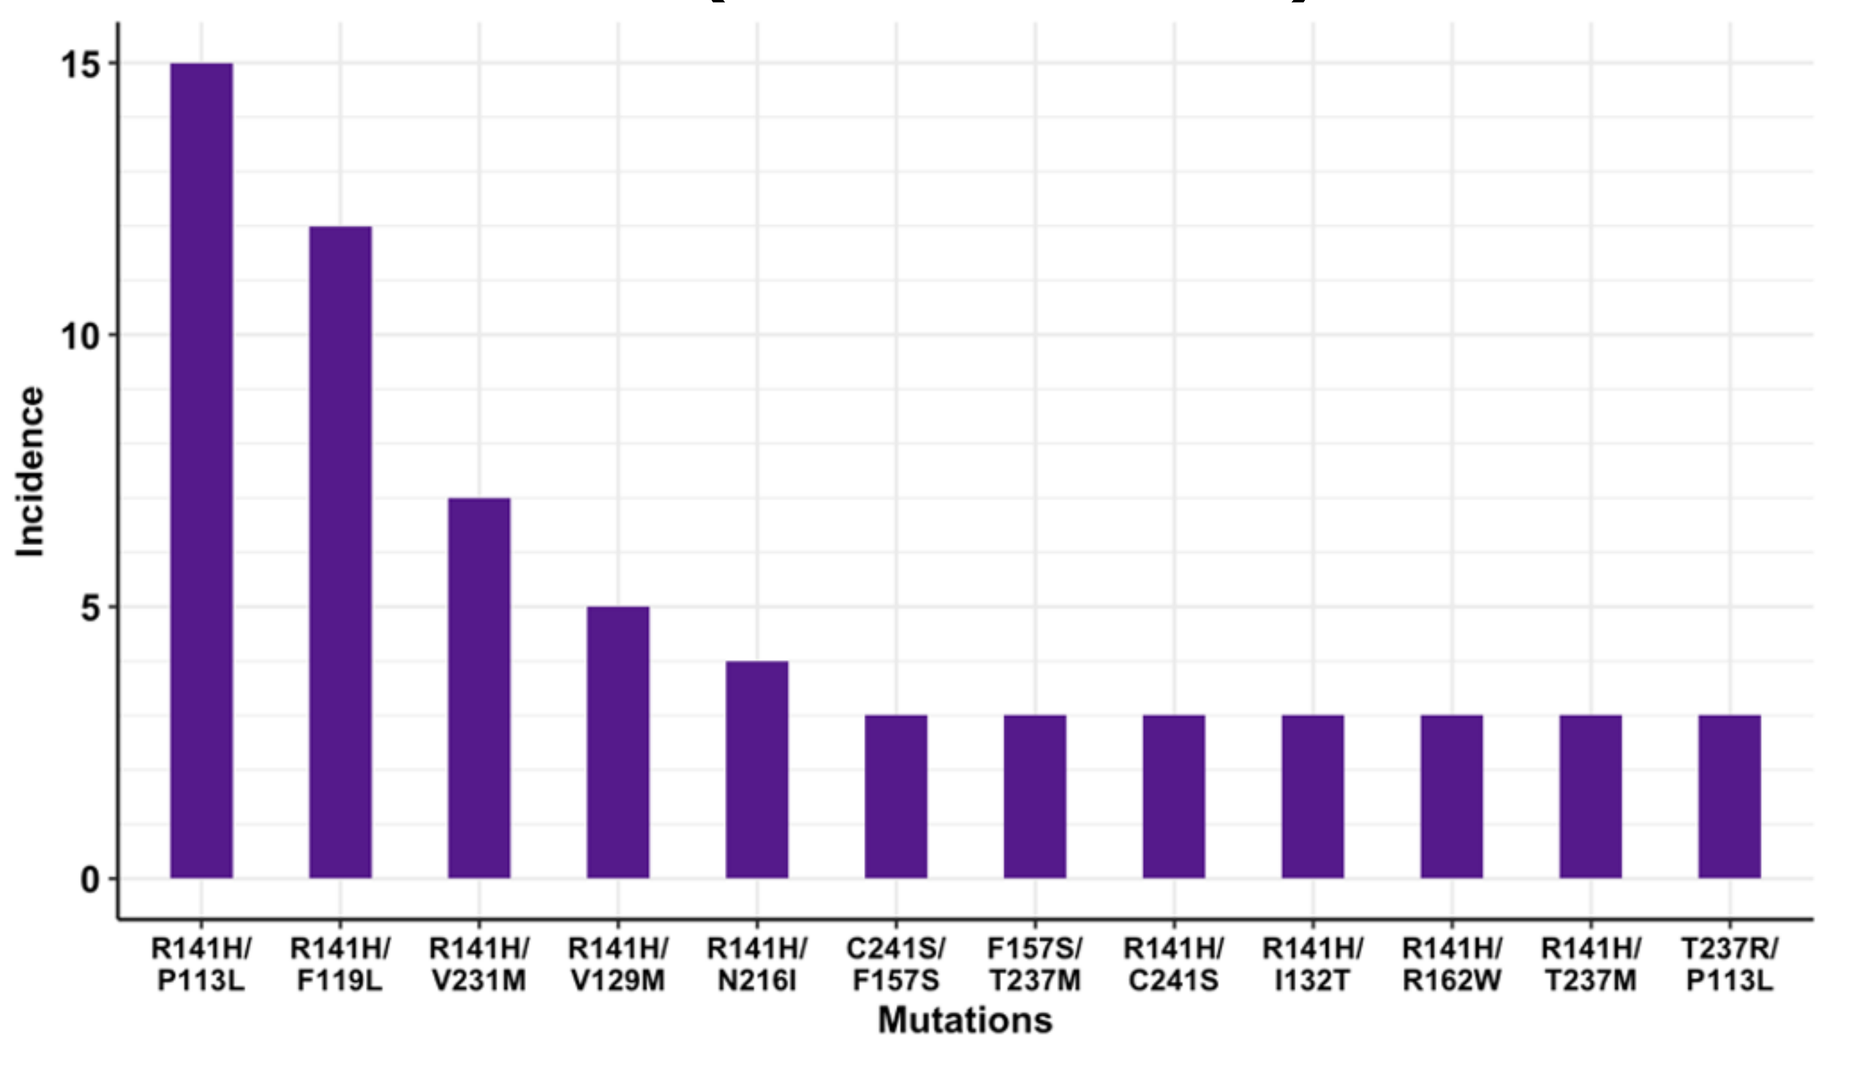

Incidence
R141H/ P113L
R141H/ F119L
R141H/ V231M
R141H/ V129M
R141H/ N216I
C241S/ F157S
F157S/ T237M
R141H/ C241S
R141H/ I132T
R141H/ R162W
R141H/ T237M
T237R/ P113L
Mutations

Failure to Thrive

A significant number of participants are below normal weight, with 49 (38%) below two z-scores (vs. 2.5% for a normal population). This is most pronounced in 0-24 months, where ~70% patients are below two z-scores. A similar pattern in seen in height, where 42 (34%) are below 2 z-scores, and this is most pronounced in adolescents (10, 53%).

Weight z-Scores at Baseline

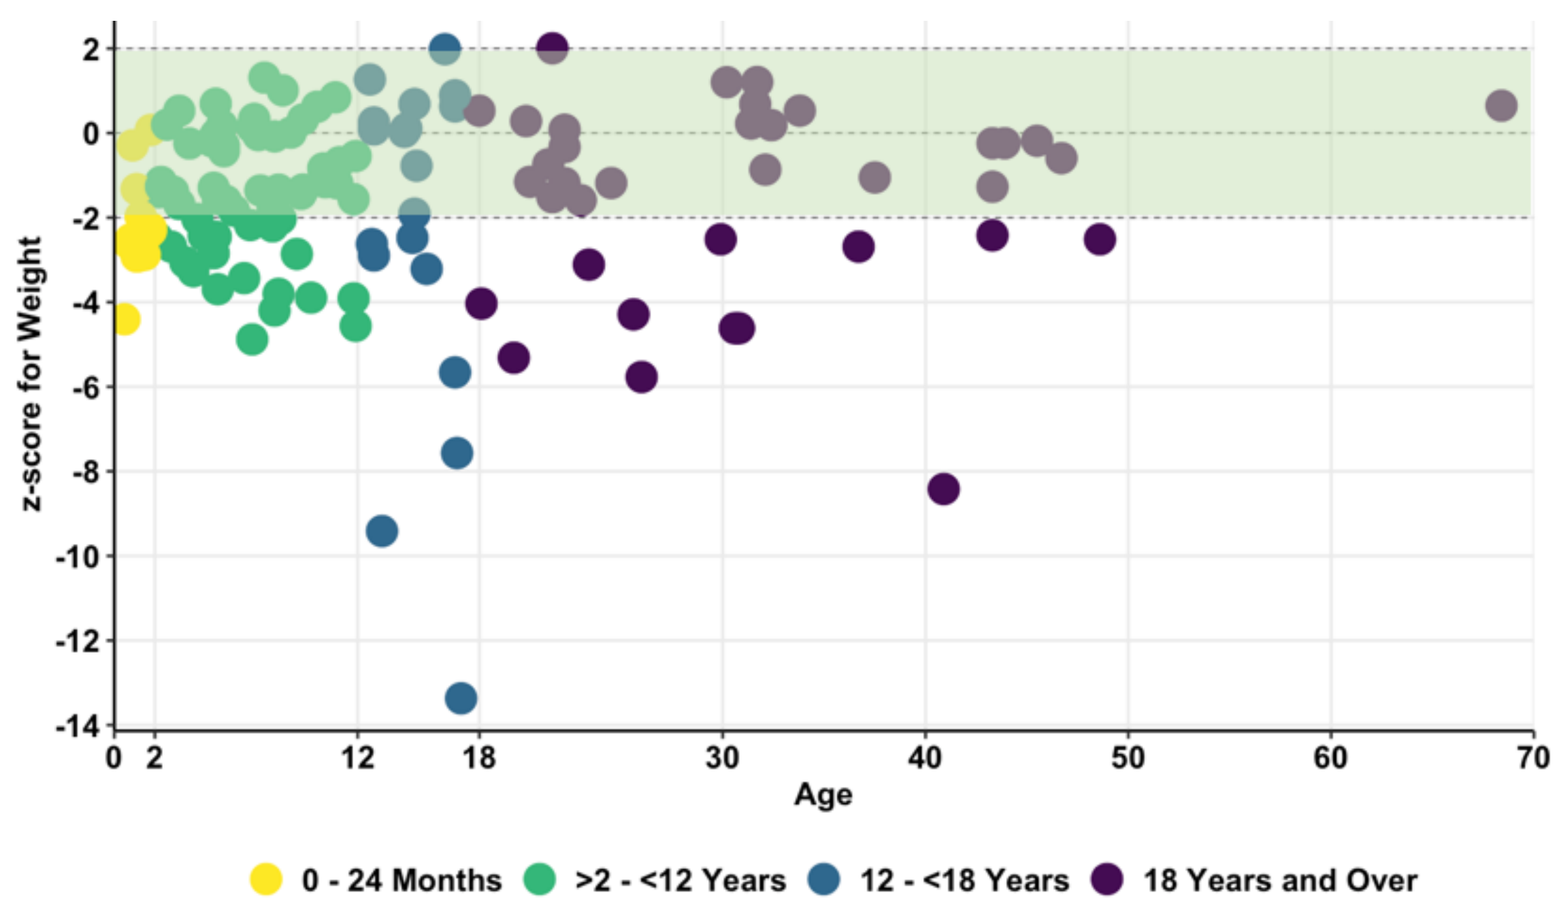

z-score for Weight
Age
0 - 24 Months
>2 - <12 Years
12 - <18 Years
18 Years and Over

N of Each Age Cohort by z-Score

Age Group	Below -2	2 to -2	Above 2
0 - 24 Months	9 (69.2%)	4 (30.8%)	0
>2 - <12 Years	20 (33.3%)	40 (66.7%)	0
12 - <18 Years	8 (42.1%)	11 (57.9%)	0
18 Years and Over	12 (32.4%)	24 (64.9%)	1 (2.7%)
All Ages	49 (38%)	79 (61.2%)	1 (0.8%)

> 50% > 40% to 50% > 30% to 40% > 20% to 30%

Coagulation

Amongst all biochemical parameters followed, coagulation parameters are one of the most consistently dysregulated, with 80% of participants below the LLN for AT activity at baseline (57% are below the LLN for both FXI and PC activity). These parameters are essentially stable over time, showing no downwards or upwards trend beyond the standard deviation in the data over the 4 year timeframe. This holds true across all age groups and for both sexes.

AT Activity at Baseline | **AT Activity Over Time**

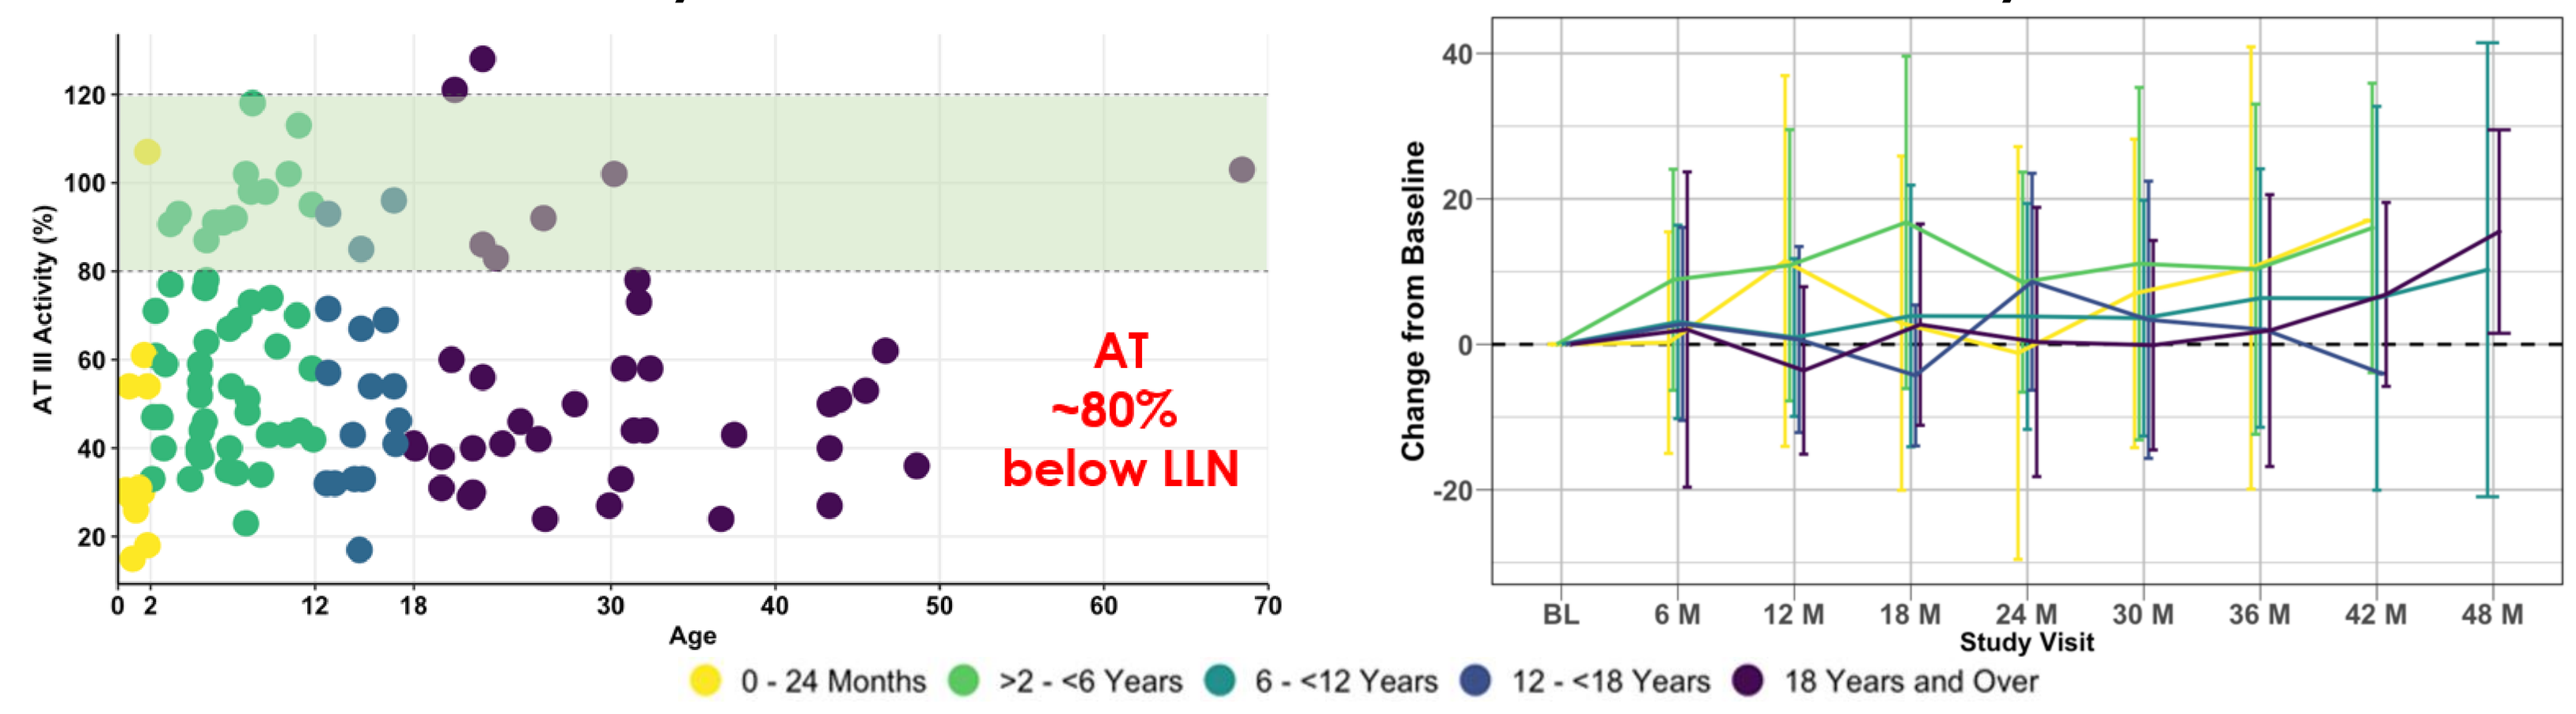

AT III Activity (%)
Age
AT
~80%
below LLN
Change from Baseline
Study Visit
0 - 24 Months
>2 - <6 Years
6 - <12 Years
12 - <18 Years
18 Years and Over

Carbohydrate Deficient Transferrin (CDT)

Most participants show a high proportion of CDT, as expected, since this evaluation is typically the first step in a diagnosis. Specifically, the asialo-/di-oligo is above the upper limit of normal for 78% of participants, while the mono-/di-oligo is for 88.1%. Similar to coagulation, there is no downwards or upwards trend beyond the standard deviation, but there is considerable visit-to-visit variability.

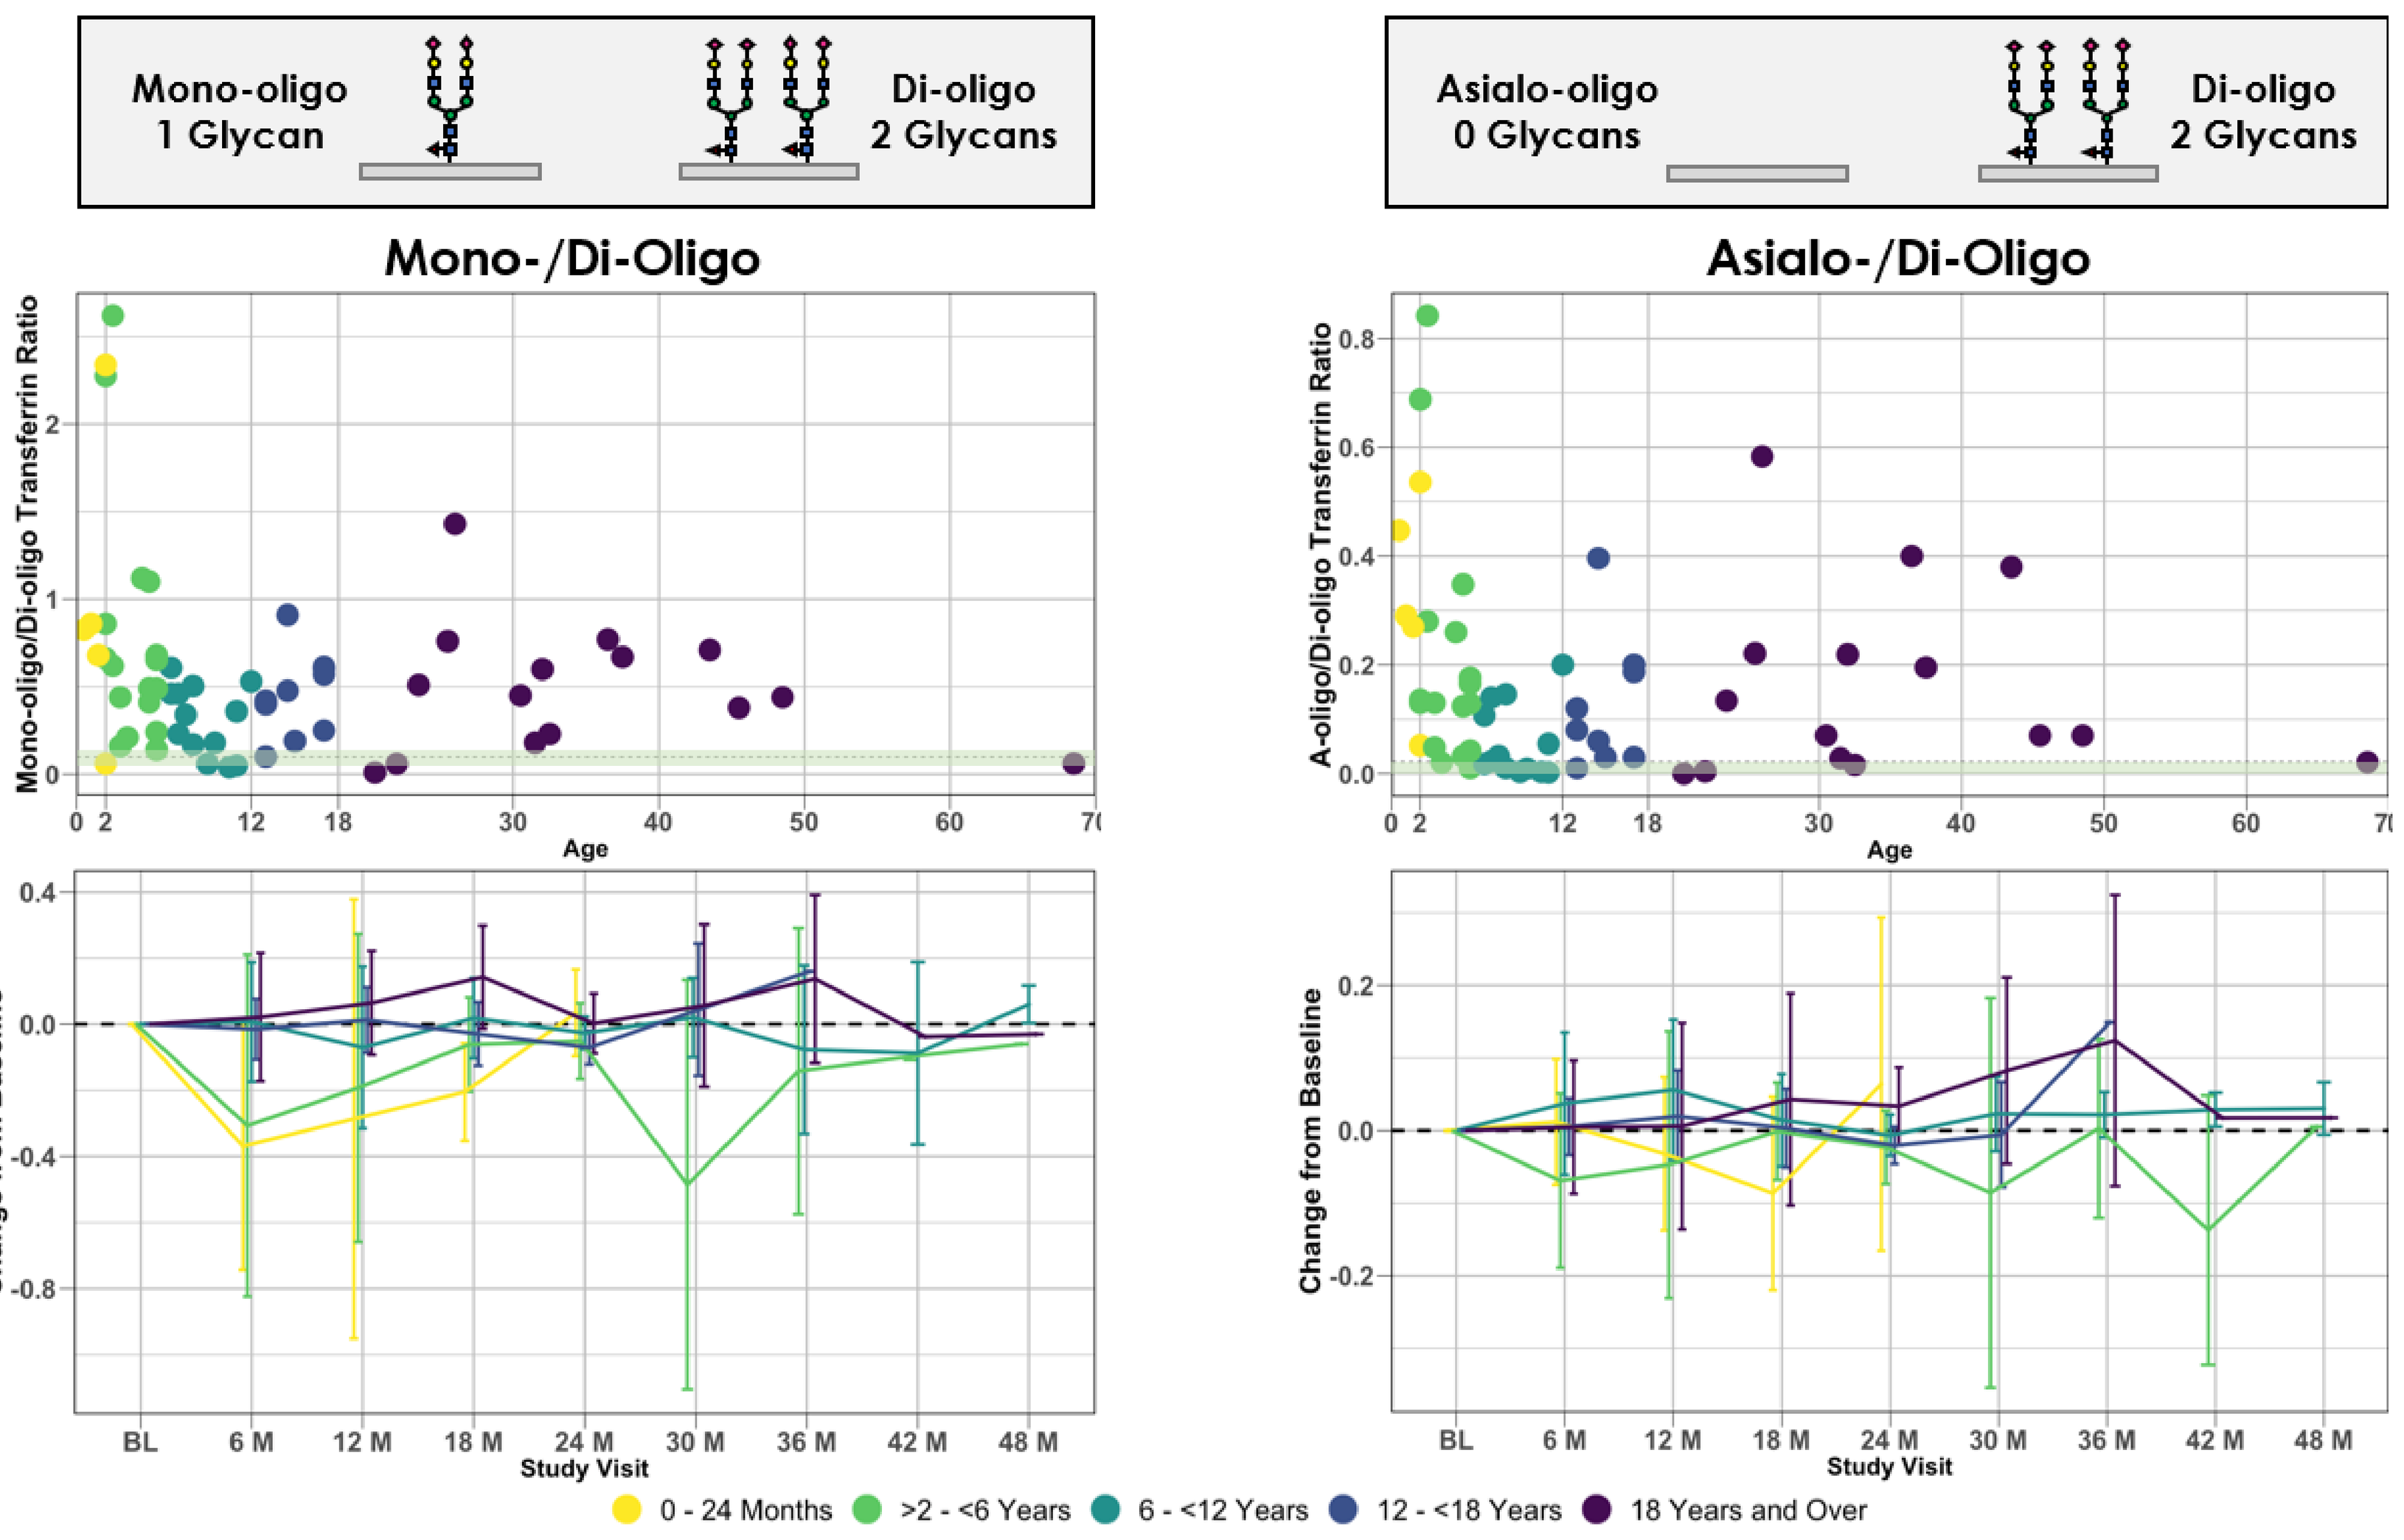

Mono-oligo
1 Glycan
Di-oligo
2 Glycans
Asialo-oligo
0 Glycans
Di-oligo
2 Glycans
Mono-/Di-Oligo
Asialo-/Di-Oligo
Mono-oligo/Di-oligo Transferrin Ratio
A-oligo/Di-oligo Transferrin Ratio
Age
Change from Baseline
Study Visit
0 - 24 Months
>2 - <6 Years
6 - <12 Years
12 - <18 Years
18 Years and Over

Liver Enzymes

Whereas coagulation and transferrin glycosylation parameters show a relatively stable (although variable) course over the 4-year timeframe of the study, the same is not true of liver parameters. Alanine transaminase (ALT) and aspartate transaminase (AST) show elevated values in childhood, especially 0 to 6 yrs, however, mostly normalize thereafter.

ALT at Baseline & Over Time | **AST at Baseline &Over Time**

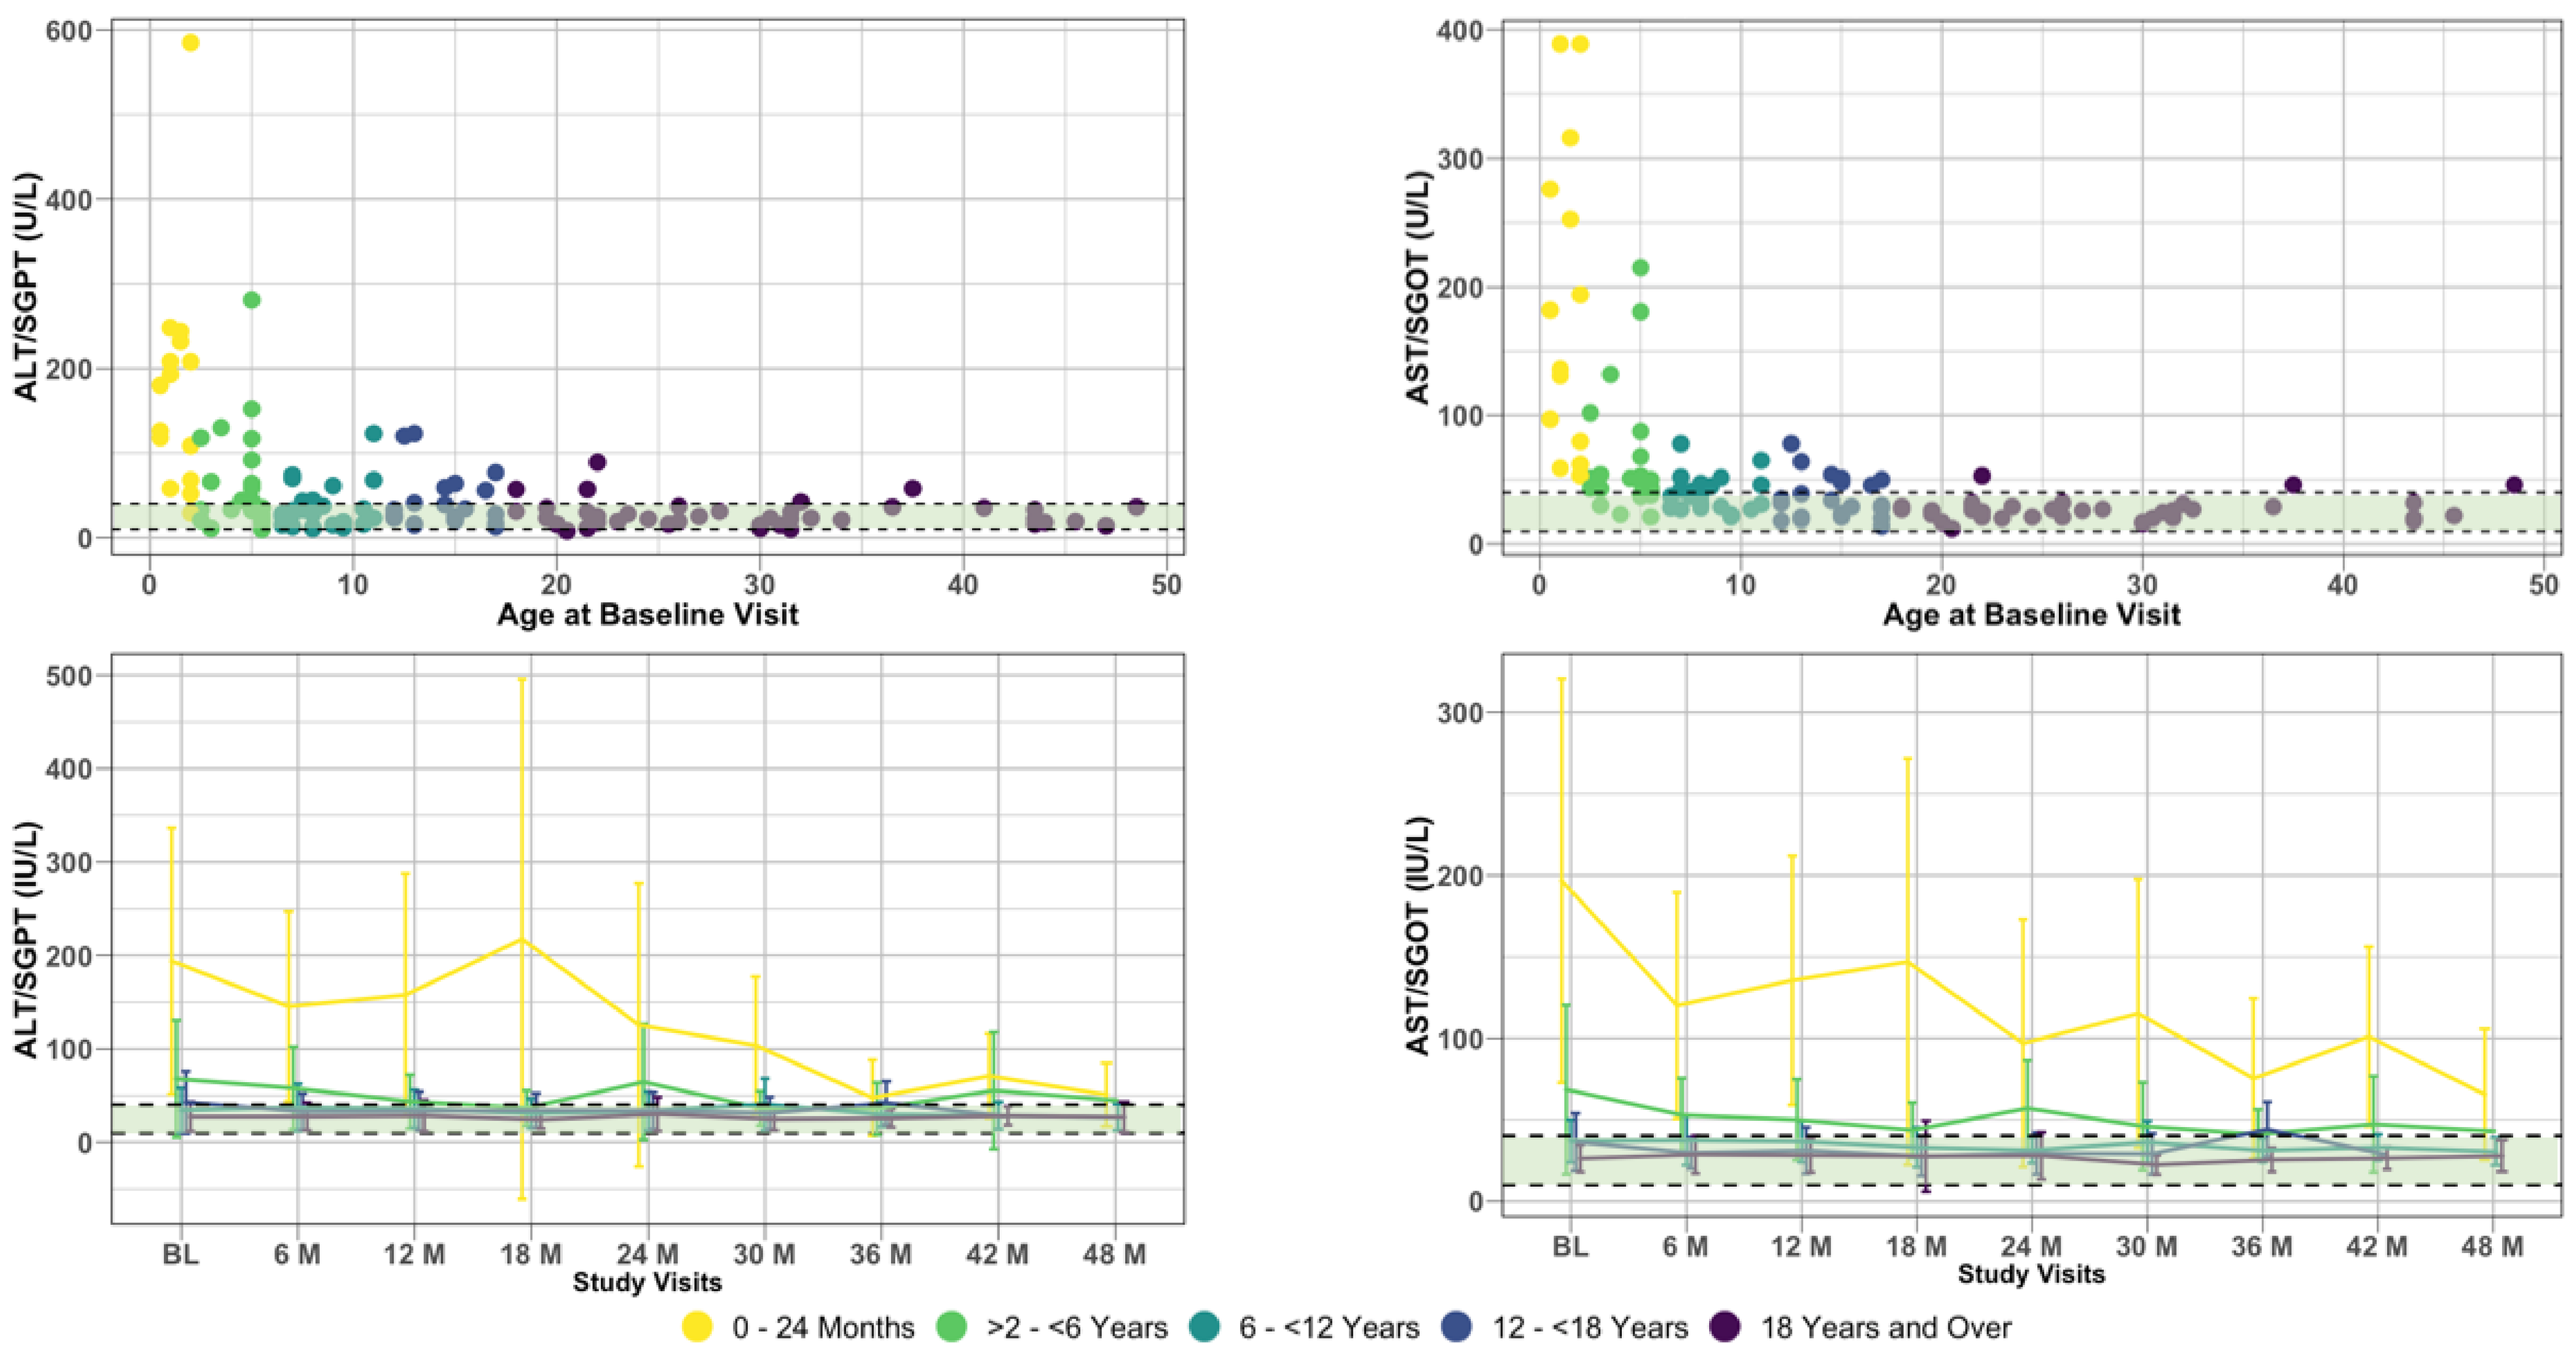

ALT/SGPT (U/L)
AST/SGOT (U/L)
Age at Baseline Visit
ALT/SGPT (IU/L)
AST/SGOT (IU/L)
Study Visits
0 - 24 Months
>2 - <6 Years
6 - <12 Years
12 - <18 Years
18 Years and Over

Adrenal Insufficiency

A significant proportion of participants were found to be at risk for adrenal insufficiency. Of 43 examined between 7am and 1pm, 11 (25.6%) were found to have both low cortisol (below 5 µg/dl) and also low to normal ACTH levels, suggestive of secondary adrenal insufficiency. This rate is significantly above the general population (2 in 10,000).

The causes are likely multifactorial; multiple enzymes and receptors in the hypothalamic-pituitary-adrenal (HPA) axis are N-glycosylated. As a result of this finding, it was recommended that monitoring of morning cortisol and ACTH levels be part of the standard care in PMM2-CDG.

ACTH vs. Cortisol

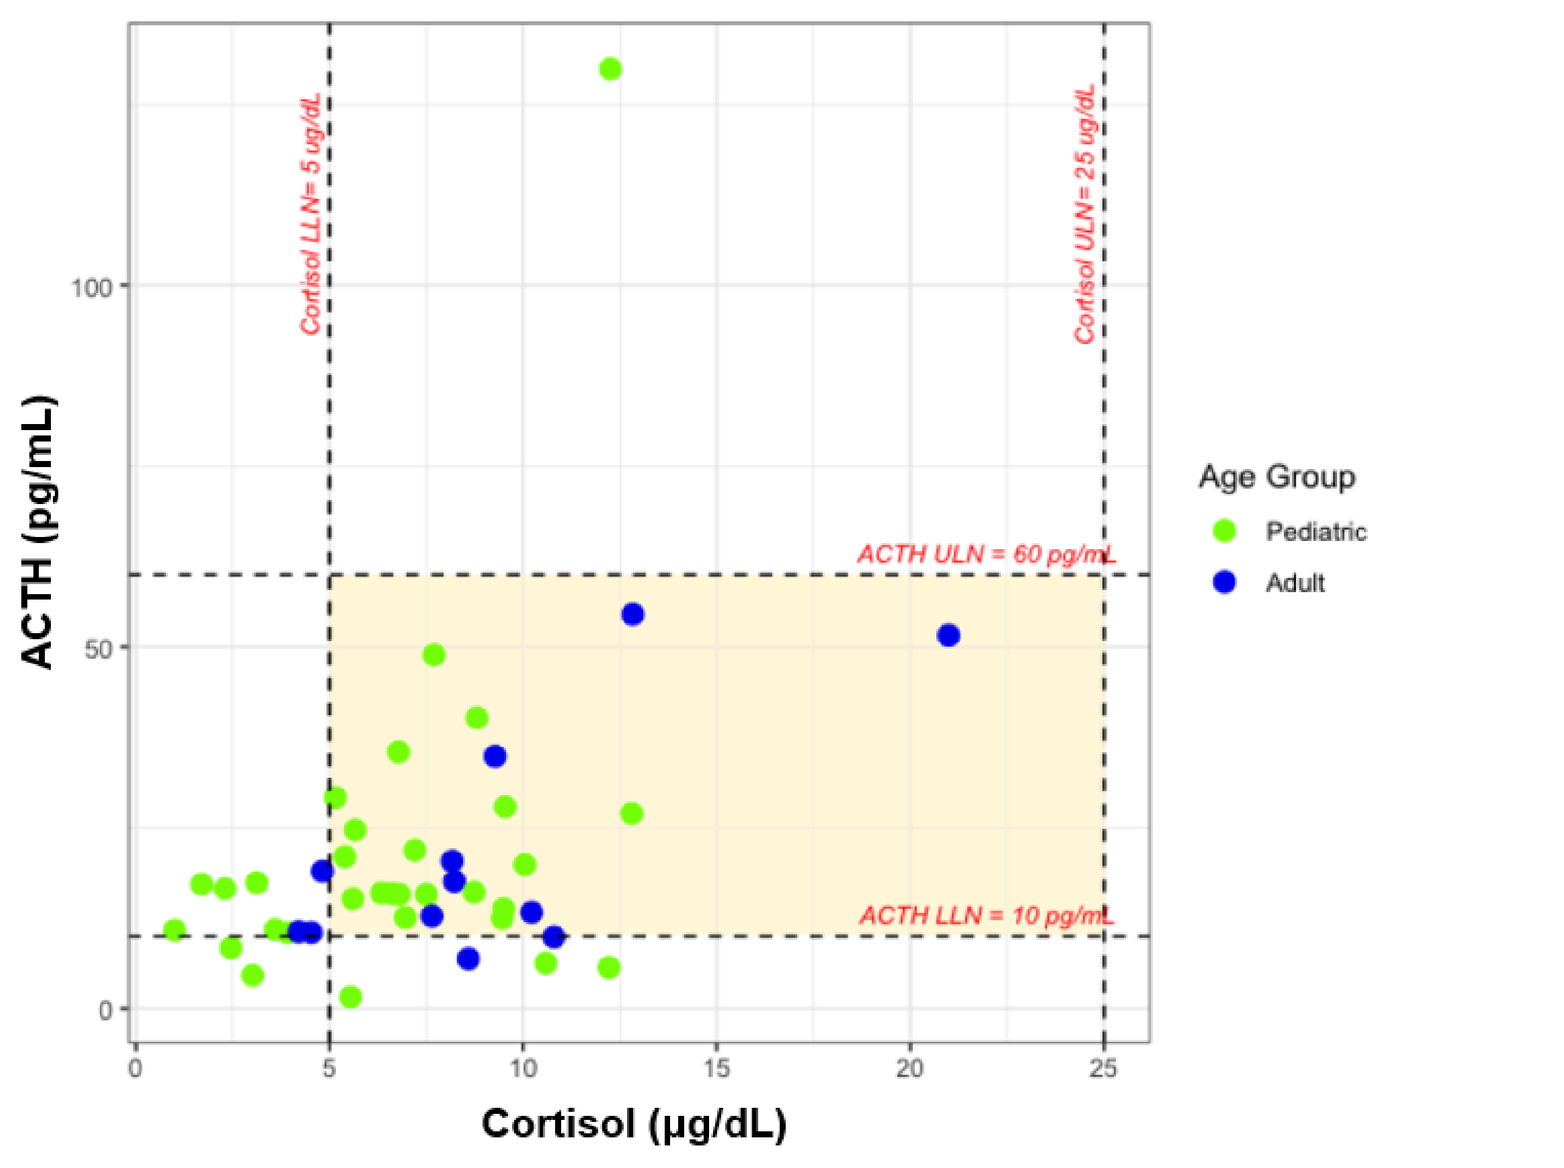

Cortisol LLN= 5 µg/dL
Cortisol ULN= 25 µg/dL
ACTH ULN = 60 pg/mL
ACTH LLN = 10 pg/mL
ACTH (pg/mL)
Cortisol (µg/dL)
Age Group
Pediatric
Adult

Conclusion

This ongoing Natural History study provides a valuable cross-sectional and longitudinal dataset to understand the clinical manifestations, morbidity, and disease progression in PMM2-CDG, which will be important for determining clinical outcome measures in future investigational human clinical trials.

RESEARCH POSTER PRESENTATION DESIGN © 2019
www.PosterPresentations.com